TOWARDS A CELL-BASED TREATMENT FOR ANDROGENETIC ALOPECIA IN MEN AND WOMEN: 12-MONTH INTERIM SAFETY RESULTS OF A PHASE I/IIA CLINICAL TRIAL USING AUTOLOGOUS DERMAL SHEATH CUP CELL INJECTIONS

RepliCel

Kevin McElwee*, Darrell Panich, David Hall, Rolf Hoffmann

RepliCel Life Sciences Inc.
Vancouver, BC, Canada
*Email: info@replicel.com

RepliCel Life Sciences is developing a patented, natural hair cell replication technology for the treatment of androgenetic alopecia (AGA) and general hair loss in men and women. This technology is comprised of a patient's own (autologous) hair follicle cells isolated from the dermal sheath cup (DSC), that have been replicated according to RepliCel's proprietary cell replication procedures. The TS001-2009 first-in-man clinical study was designed to test the safety and efficacy of RepliCel's technology in 10 men and 10 women with AGA. The trial protocol, was developed in compliance with International Conference on Harmonisation guidelines for Good Clinical Practice (ICH GCP) based on advice received from the Paul-Ehrlich-Institut; an Agency of the German Federal Ministry of Health. Before the study was initiated at the National Scientific Research Centre for Skin and Venereal Diseases in Tbilisi, Georgia, the protocol received a thorough scientific and ethical review by the Georgian National Council of Bioethics which approved the conduct of the study on Oct, 27, 2010.

CLINICAL TRIAL PROTOCOL

The **primary objective** of the study is to assess the local (at treatment sites) safety profile of injections of autologous dermal sheath cup cells (DSCC) at 6 months post-injection compared to control. **Secondary objectives** were to assess systemic (overall) safety, local safety, and efficacy at 24 months post-injection.

SCREENING

As part of the screening process subjects had their mild to moderate AGA categorized either according to the Ludwig Scale (female) or the Norwood scale (male) and provided blood samples to confirm their health status (i.e. to exclude HIV, HBV and HCV). Once the participants' health status was confirmed, 2 cm² treatment sites selected on either side of the fronto-parietal scalp region and occipital scalp biopsies were obtained for manufacturing of the cell therapy product.

TREATMENT

Once manufacturing was completed, participants returned to the clinic to receive randomized, blinded injections of autologous replicated DSCC in a carrier medium (verum) in one pre-selected treatment site, and of carrier medium alone (control) in the other treatment site. A proprietary syringe holder was designed to perform injections for the study in a consistent fashion. Each 2 cm² treatment site received a total of 6 injections that delivered 1.0ml of either verum or control product. Distribution of the 6 injections was controlled by an injection template; indicating orientation of the syringe holder for each injection. Depth and angle of each injection was controlled by the design of the holder itself. Rather than injecting products as a bolus, they were released as the needle was retracted from the scalp; thereby leaving a line of product at various levels within the dermis. This ensured that cells came in contact with both the hair follicles and the basal layer of the epidermis.

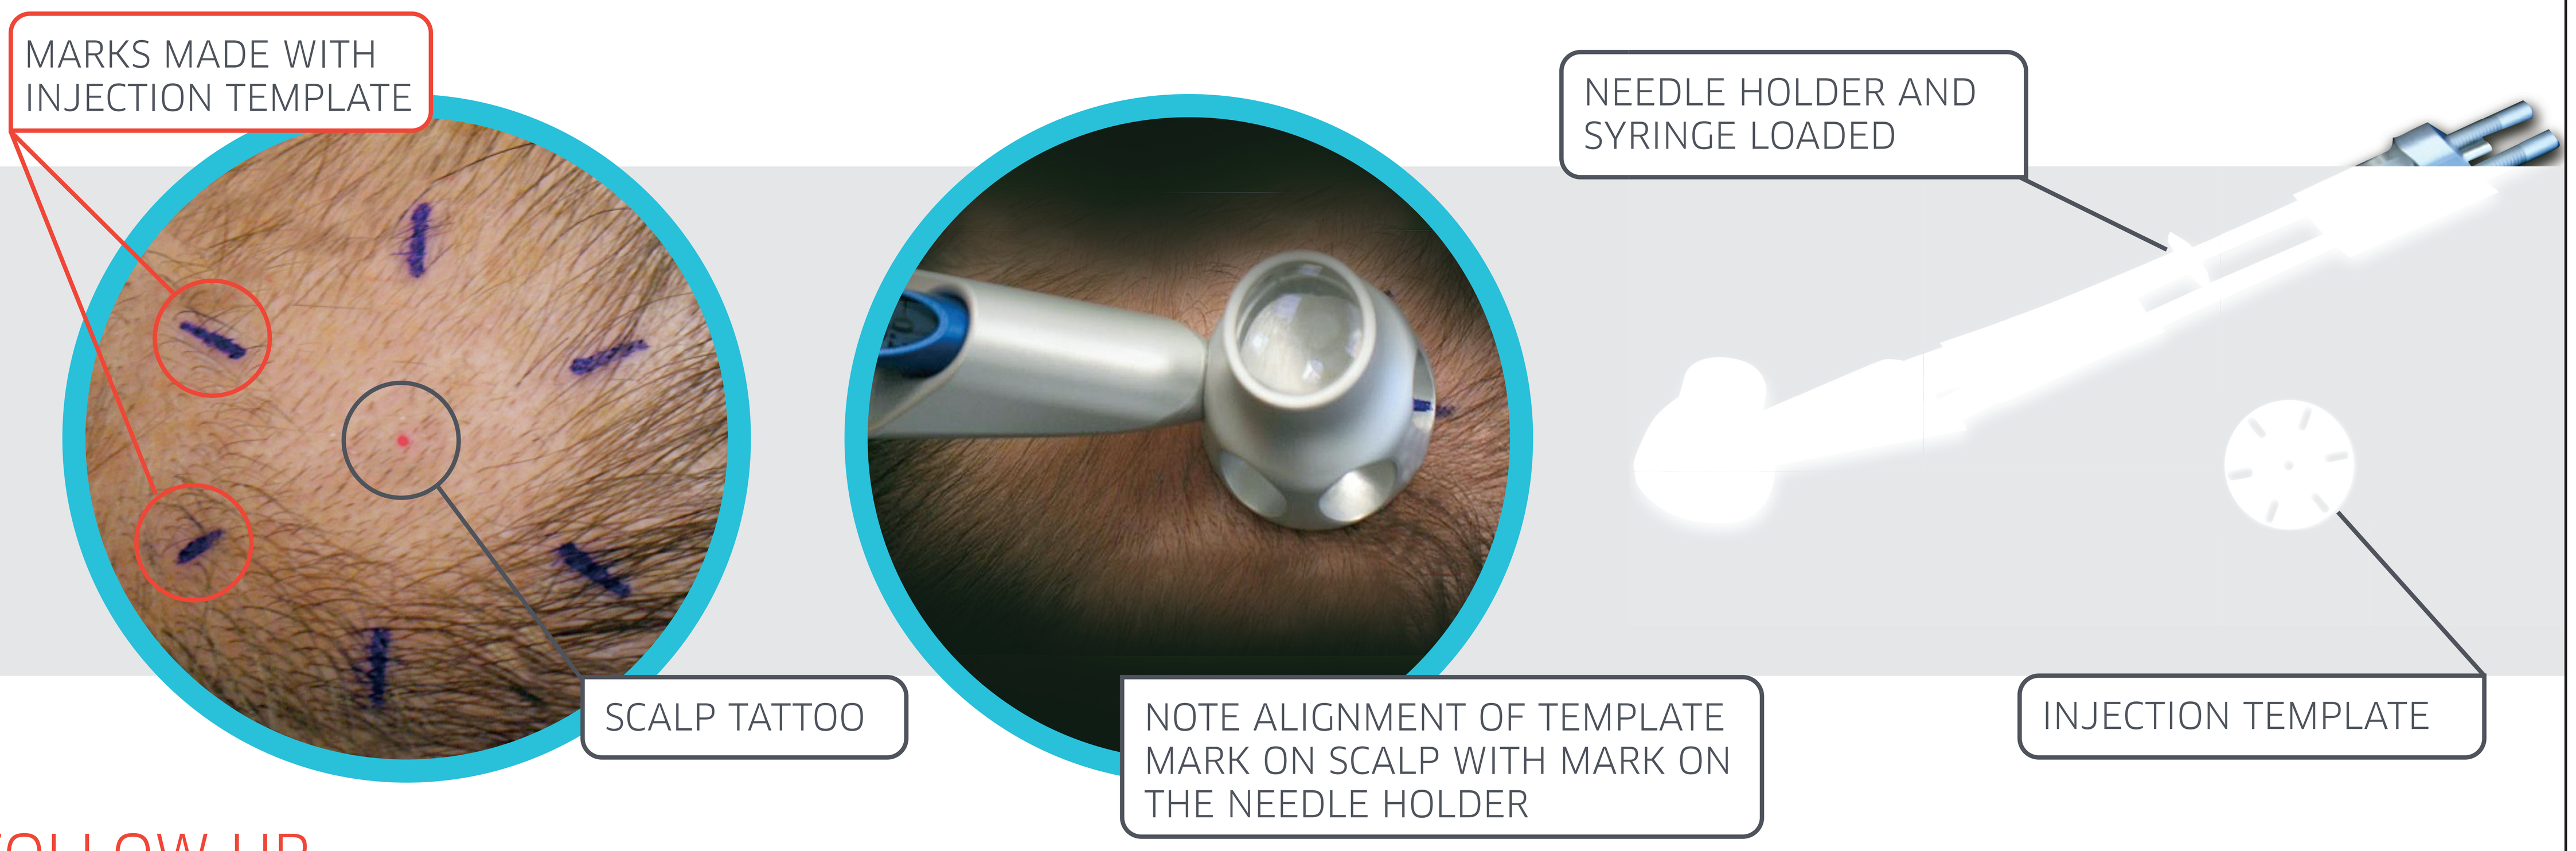

MARKS MADE WITH INJECTION TEMPLATE
NEEDLE HOLDER AND SYRINGE LOADED
SCALP TATTOO
NOTE ALIGNMENT OF TEMPLATE MARK ON SCALP WITH MARK ON THE NEEDLE HOLDER
INJECTION TEMPLATE

FOLLOW UP

During the post-injection follow-up period, participants returned to the clinic for ten follow-up visits over a 24-month period to have their health closely monitored to ensure that there were no adverse effects associated with receiving the injections and to determine the efficacy of hair follicle cell injections at stimulating hair growth. Local safety was assessed through reports from study participants, observations made by the study investigator and through assessment of treatment site images by independent investigators. Systemic safety was assessed through reports of adverse events by participants and observations of the study investigator. Furthermore, 4 subjects (2 male, 2 female) were selected at random to provide biopsies from the treatment sites at the 6- and 12-month post injection time point. An additional 4 patients will provide biopsies at the 24-month time-point. These biopsies were sent to an independent laboratory for histopathological analysis. Efficacy was assessed through analysis of images of treatment sites made with TrichoScan® equipment and software.

INTERIM ANALYSES AT 6- AND 12-MONTHS

Once all participants completed their 12-month post-injection follow-up visit, all safety and efficacy data was collected for analysis. Results from this analysis were performed to allow for an accurate assessment of patient safety issues and to provide information to assist in the design of future clinical trials. Additionally, RepliCel analyzed early efficacy at 6-months.

Patient recruitment

- Screened 30 subjects (16 male, 14 female).
- Harvested 25 biopsies (12 male, 13 female).
- 19 subjects injected (10 male, 9 female).
- 3 subjects had to be excluded from efficacy result analysis because their injection products were shipped outside of the temperature range stated in the protocol.

RESULTS

Local Tolerance (Safety) @ 12-months post injection

- The majority of injections were well-tolerated.
- 6 (32%) participants experienced a local reaction (mild burning) related to injections of autologous DSCC (verum) while 3 (16%) experienced local reactions associated with injections of control (burning, pyoderma, excoriation).
- All reactions associated with injections of verum were reported within the first 3 days post-injection and were resolved within 24 hrs.
- Histopathological analysis of biopsies taken from 4 participants at 6-months post-injection and 12-months post-injection (8 patients, 16 biopsies in total) revealed no pathology suggestive of a tumour, granuloma, or hamartoma.

Systemic Safety

- No serious adverse events reported in first 12 months post-injection.
- None of the 30 events reported by 13 subjects were considered related to study injections.

Early Efficacy at 6 months post-injection

- Significantly more subjects demonstrated a 5% increase in hair density at verum-treated sites vs. control-treated sites (63% vs. 37%).
- 63% of the verum-treated subjects (10 of 16) had 5% increase in total hair density.
- 70% of those 10 subjects had 10% density growth.
 - Including: 19.6%, 19.2%, 17.2%, 13.6%, 12.0%, 11.2% & 10.6%
 - Average = 14.2% for the 7 of 10 subjects 10%
 - Average = 11.8% for the 10 of 10 subjects 5%

Phase I — First-Man-In

6 Month Post Injection Image from One Patient

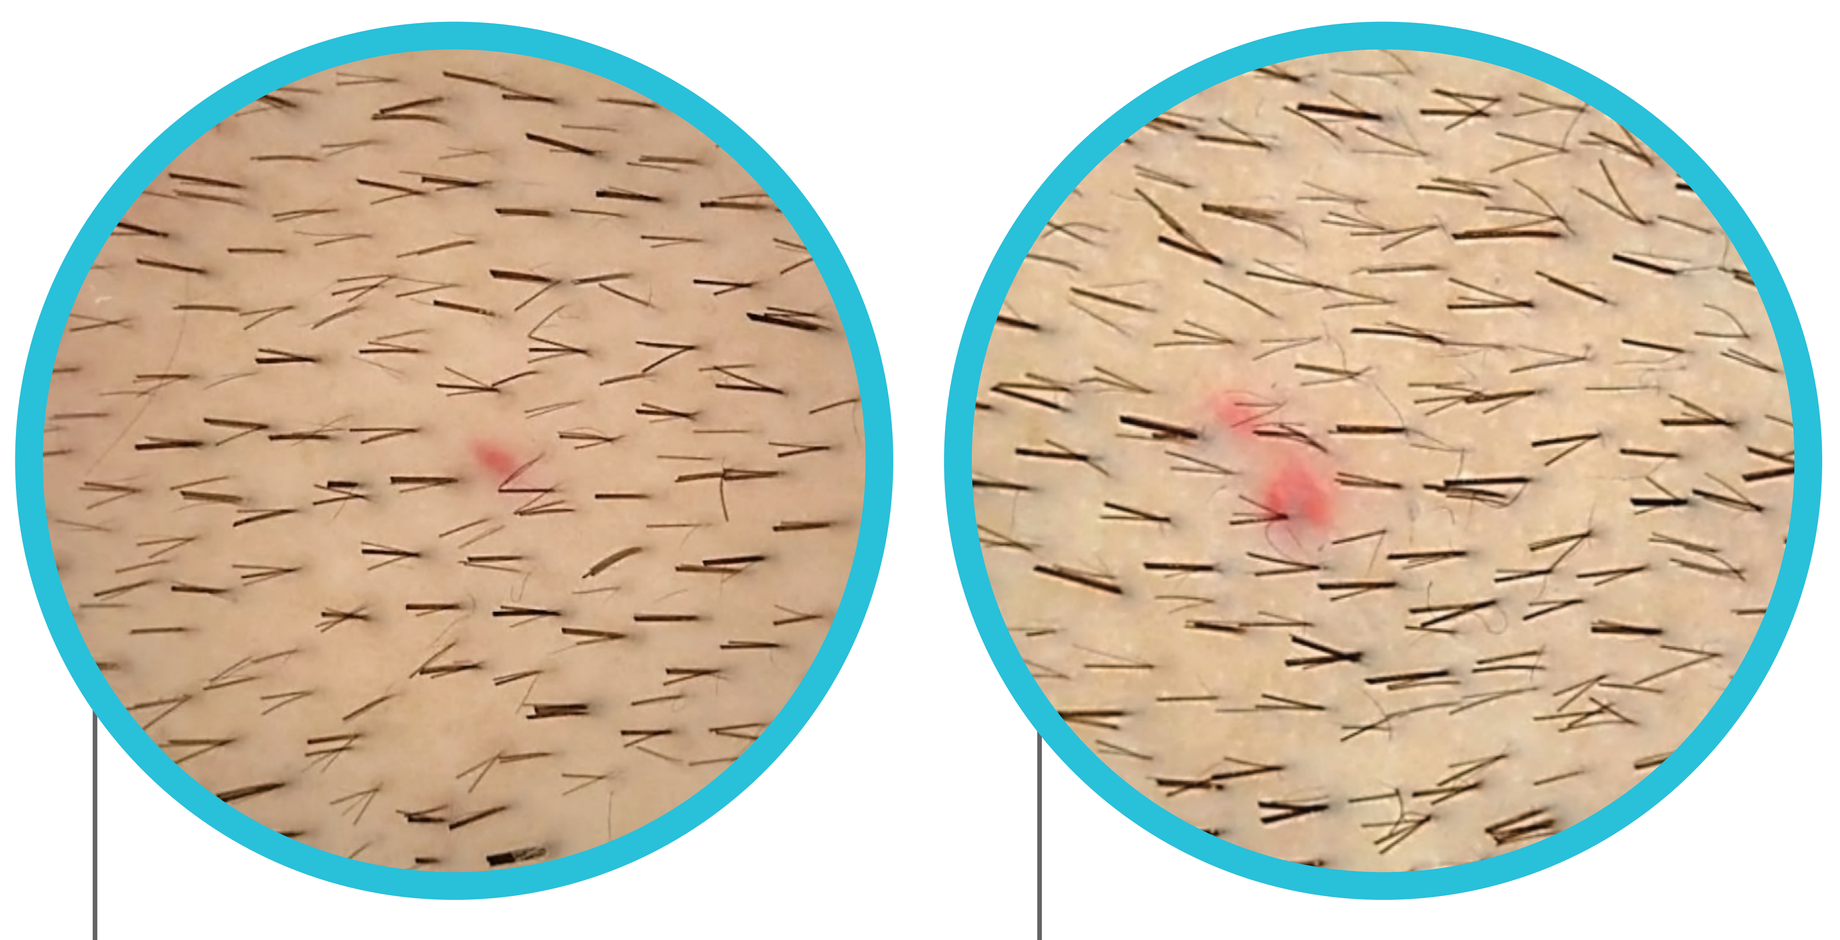

Visit 1 Pre-Injection (Baseline)

Visit 7 6 Months Post-Injections

- 24.9% Increase in vellus hair
- 14.5% Increase in terminal hair
- 19.2% Total increase in hair density
- 15.4% Increase of cumulative thickness per area

Tattoo refreshed at 6 months

Phase I — Six Month Interim Results

Secondary Interim Endpoint - Early Efficacy

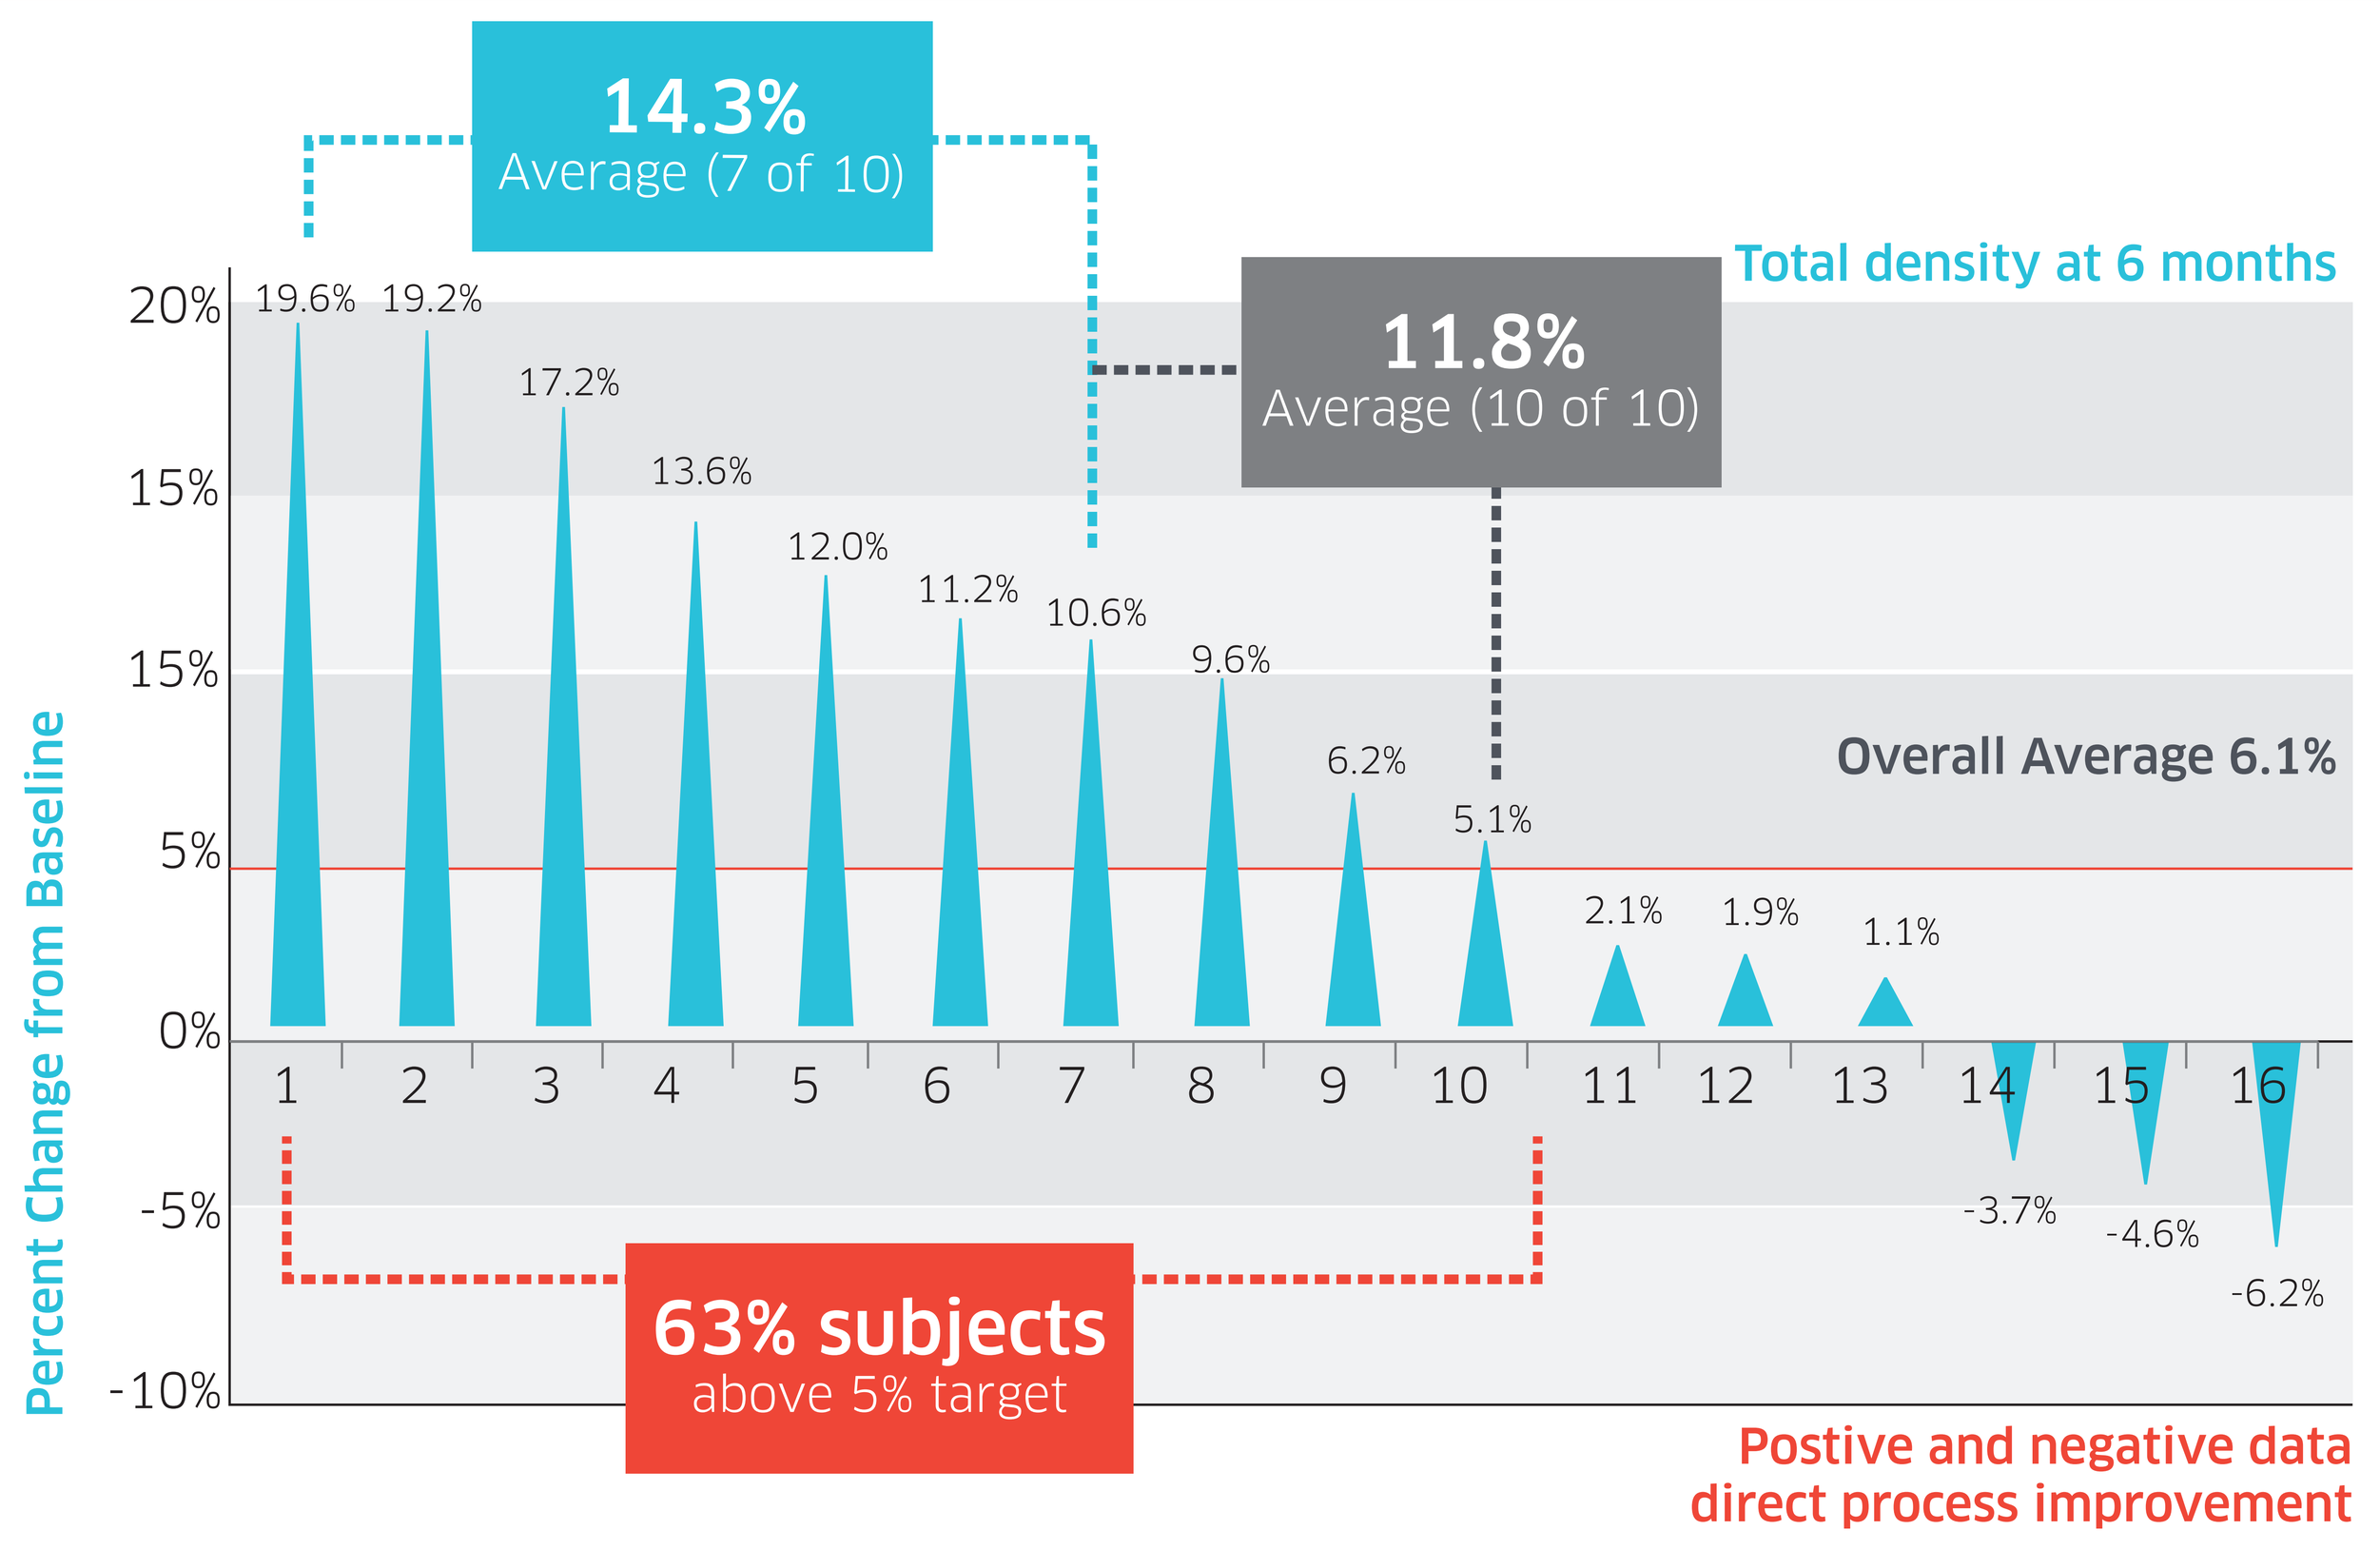

14.3%
Average (7 of 10)
11.8%
Average (10 of 10)
Total density at 6 months
Overall Average 6.1%
Percent Change from Baseline
20%
15%
15%
5%
0%
-5%
-10%
19.6% 19.2% 17.2% 13.6% 12.0% 11.2% 10.6% 9.6% 6.2% 5.1% 2.1% 1.9% 1.1% -3.7% -4.6% -6.2%
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16
63% subjects
above 5% target
Postive and negative data
direct process improvement

CONCLUSION

The safety data collected from study participants 12-months after receipt of injections of verum and control revealed that the RepliCel™ treatment is very safe and well-tolerated. This data, coupled with positive increases in hair growth efficacy measured at 6-months post-injection, provide support for the Company to move forward with development of a Phase II dosing trial. RepliCel is currently working with European regulators to finalize an application to conduct this Phase II trial, expected to commence in late 2013. This trial will be designed to optimize the regimen utilized for the treatment of AGA through evaluation of several different concentrations of cells and treatment schedules.

www.replicel.com