Autologous Cell Treatment for Achilles Tendinosis: A Phase I/II Clinical Trial

Rainer Marksteiner[2], Birte Magnus[2], Hisae Nakamura[1], Lee Buckler[1], Kevin J McElwee[1], Darell Panich[1], David Hall[1] and Rolf Hoffmann[1]

[1]RepliCel Life Sciences Inc., Vancouver, Canada

[2]Innovacell Biotechnologie AG, Innsbruck, Austria

Introduction and Objectives

Tendinosis is the most prevalent form of overuse tendon injuries, impairing 30 to 50 million people worldwide. Tendinosis results from degeneration of tendon cells and collagen fibres caused by repetitive tendon usage and aging. Currently available treatments for tendinosis do not often mediate complete recovery and leave individuals immobilized for several months. Cell-based regenerative medicine therapies have generated considerable amount of research interest in the recent years and could provide great potential in treating chronic conditions, such as tendinosis.

We have developed a tissue-engineered product containing autologous cells capable of producing type I collagen, which plays an important role in maintaining a healthy tendon. The objectives of our studies were to evaluate pre-clinical safety and efficacy data prior to conducting a clinical trial.

Methods and Results

To obtain collagen-producing cells, the non-bulbar dermal sheath (NBDS) cells were isolated from the hair follicles of healthy individuals and cultured using serum and growth factor supplemented fibroblast medium (Figure 1). To investigate if NBDS cells produce functional proteins and respond to mechanical force, NBDS cells cultured and mixed in autologous plasma, and a resulting gel-like structure containing cells was placed under a tensile force *in vitro.* Similarly, dermal fibroblasts (FB) and tendon cells were separately cultured, and the expression of tendon-related proteins was compared. Results (Figure 2) showed that a similar protein expression profile between NBDS cells, tendon derived cells and dermal fibroblasts, reflecting their abilities to produce tendon related proteins under mechanical force (stretching).

To evaluate safety, we have conducted GLP-compliant tolerance, tumorigenicity and biodistribution studies using rabbits and immune deficient mice. The results showed that NBDS cells were well-tolerated and did not form tumors or migrate to secondary sites (Table 1).

Figure 1. Schematic depiction of DSC cell location

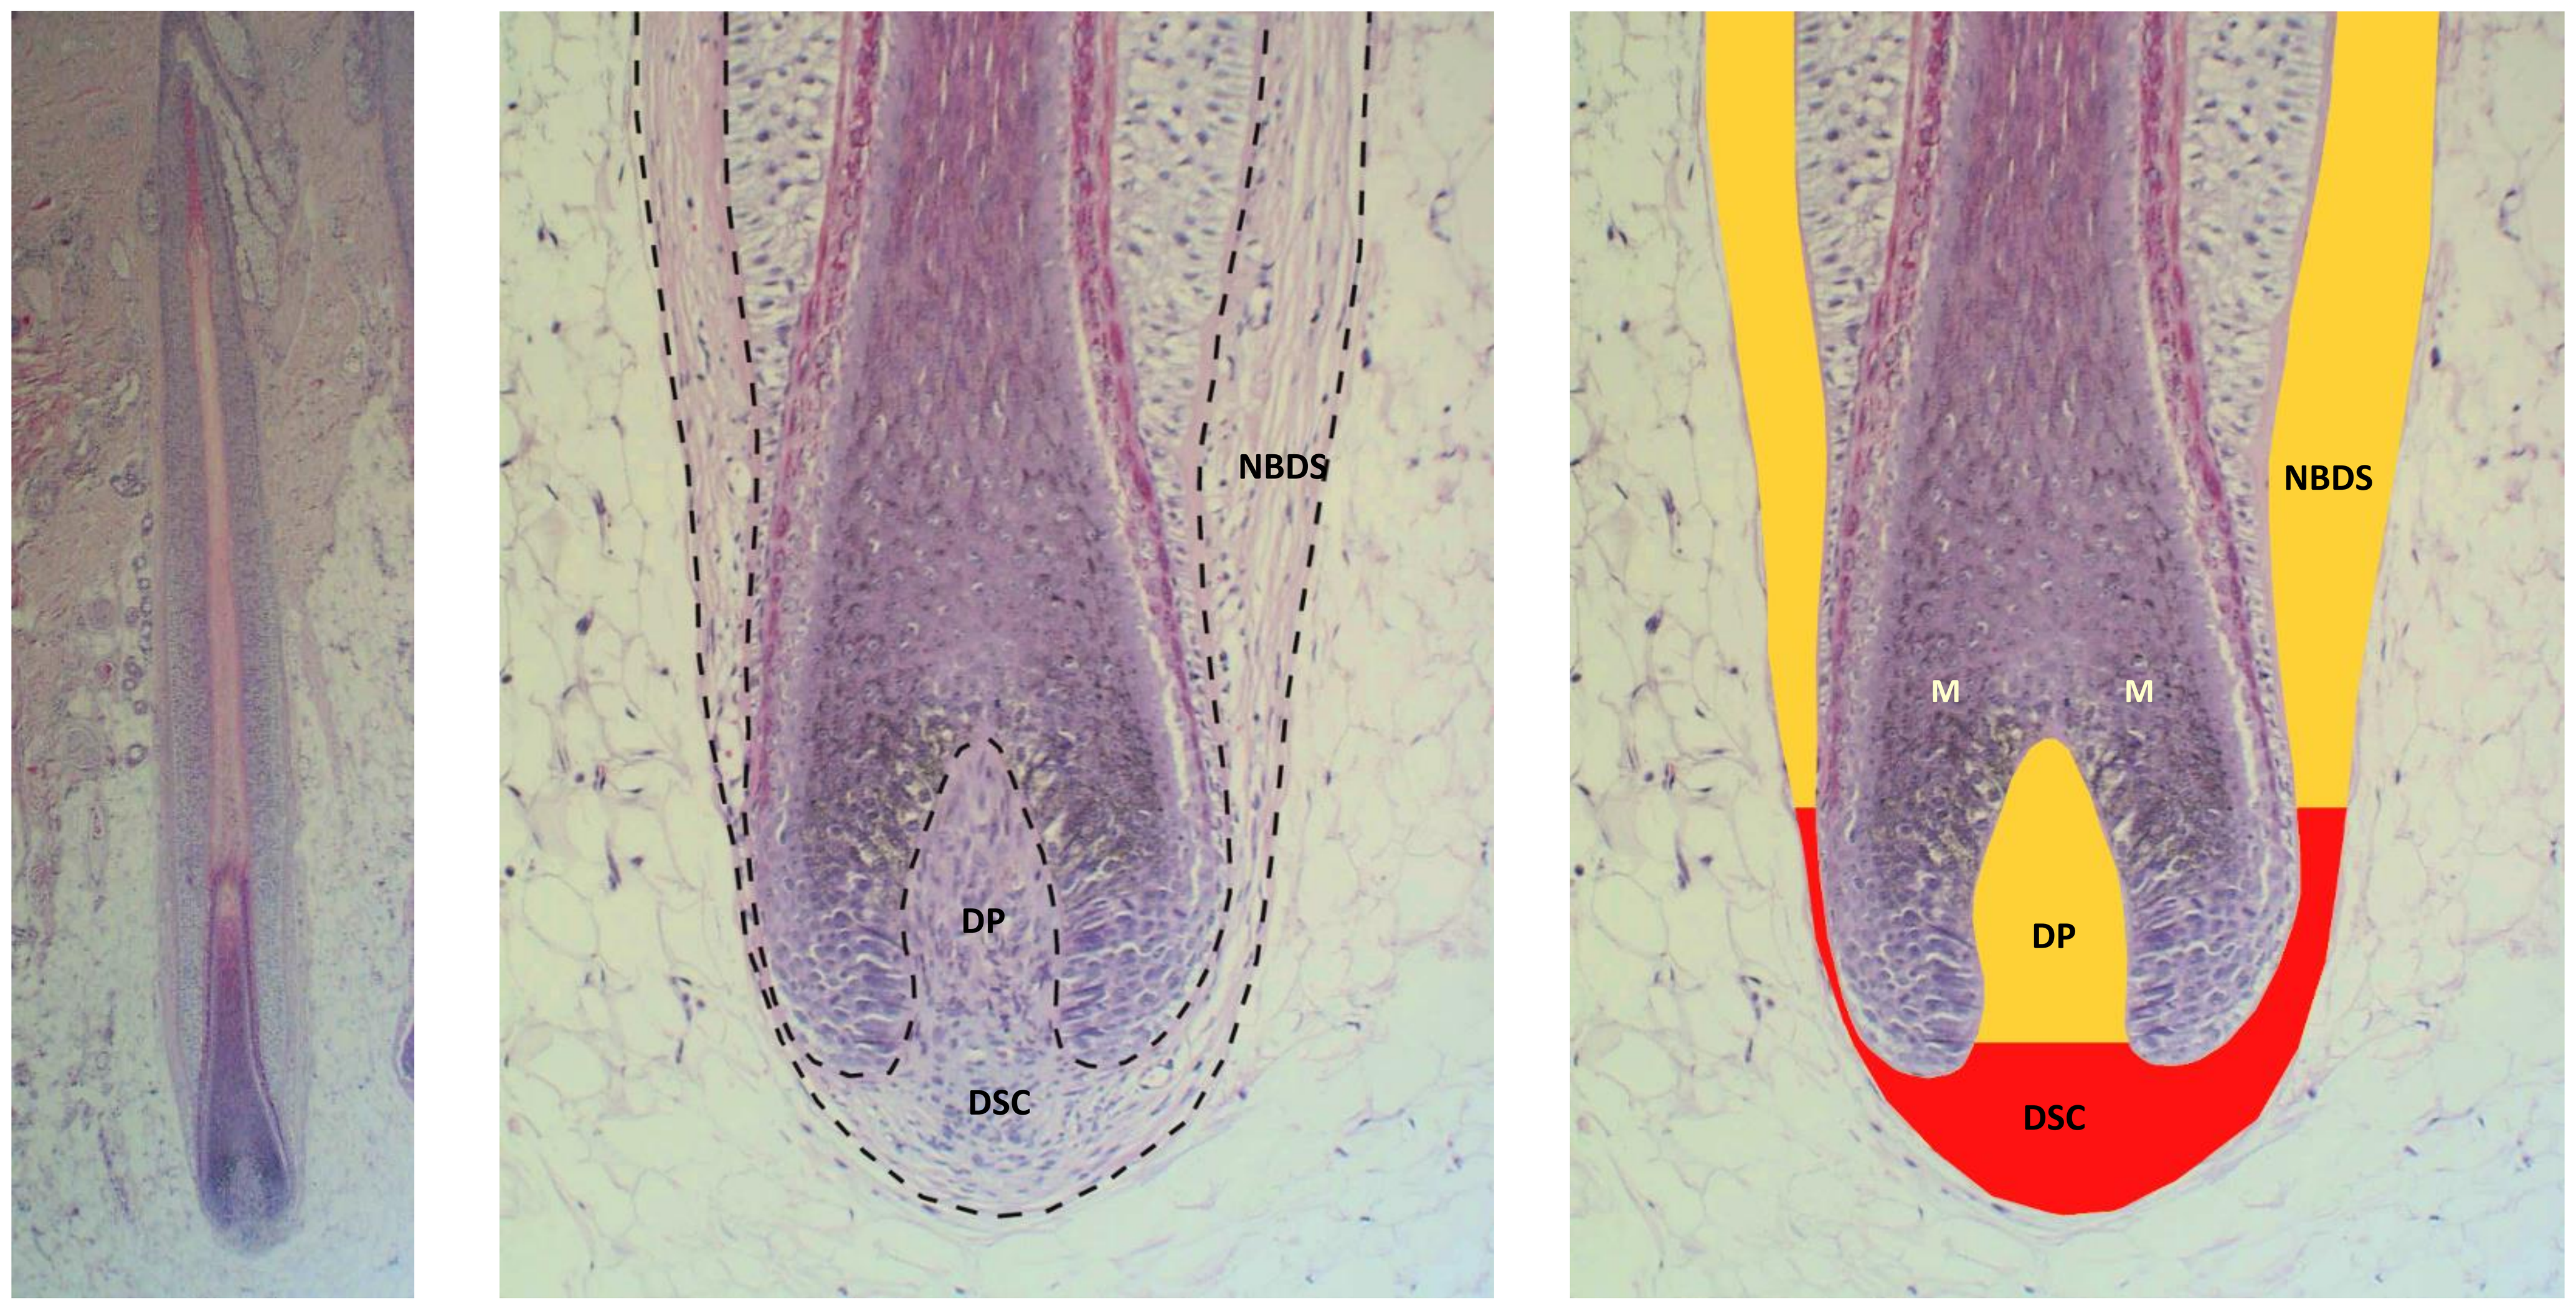

NBDS
DP
DSC
NBDS
M
M
DP
DSC

DP – dermal papilla
DSC – dermal sheath cup
NBDS – non-bulbar dermal sheath
M – hair follicle matrix cells

Figure 2. Histological staining of tendon-related proteins

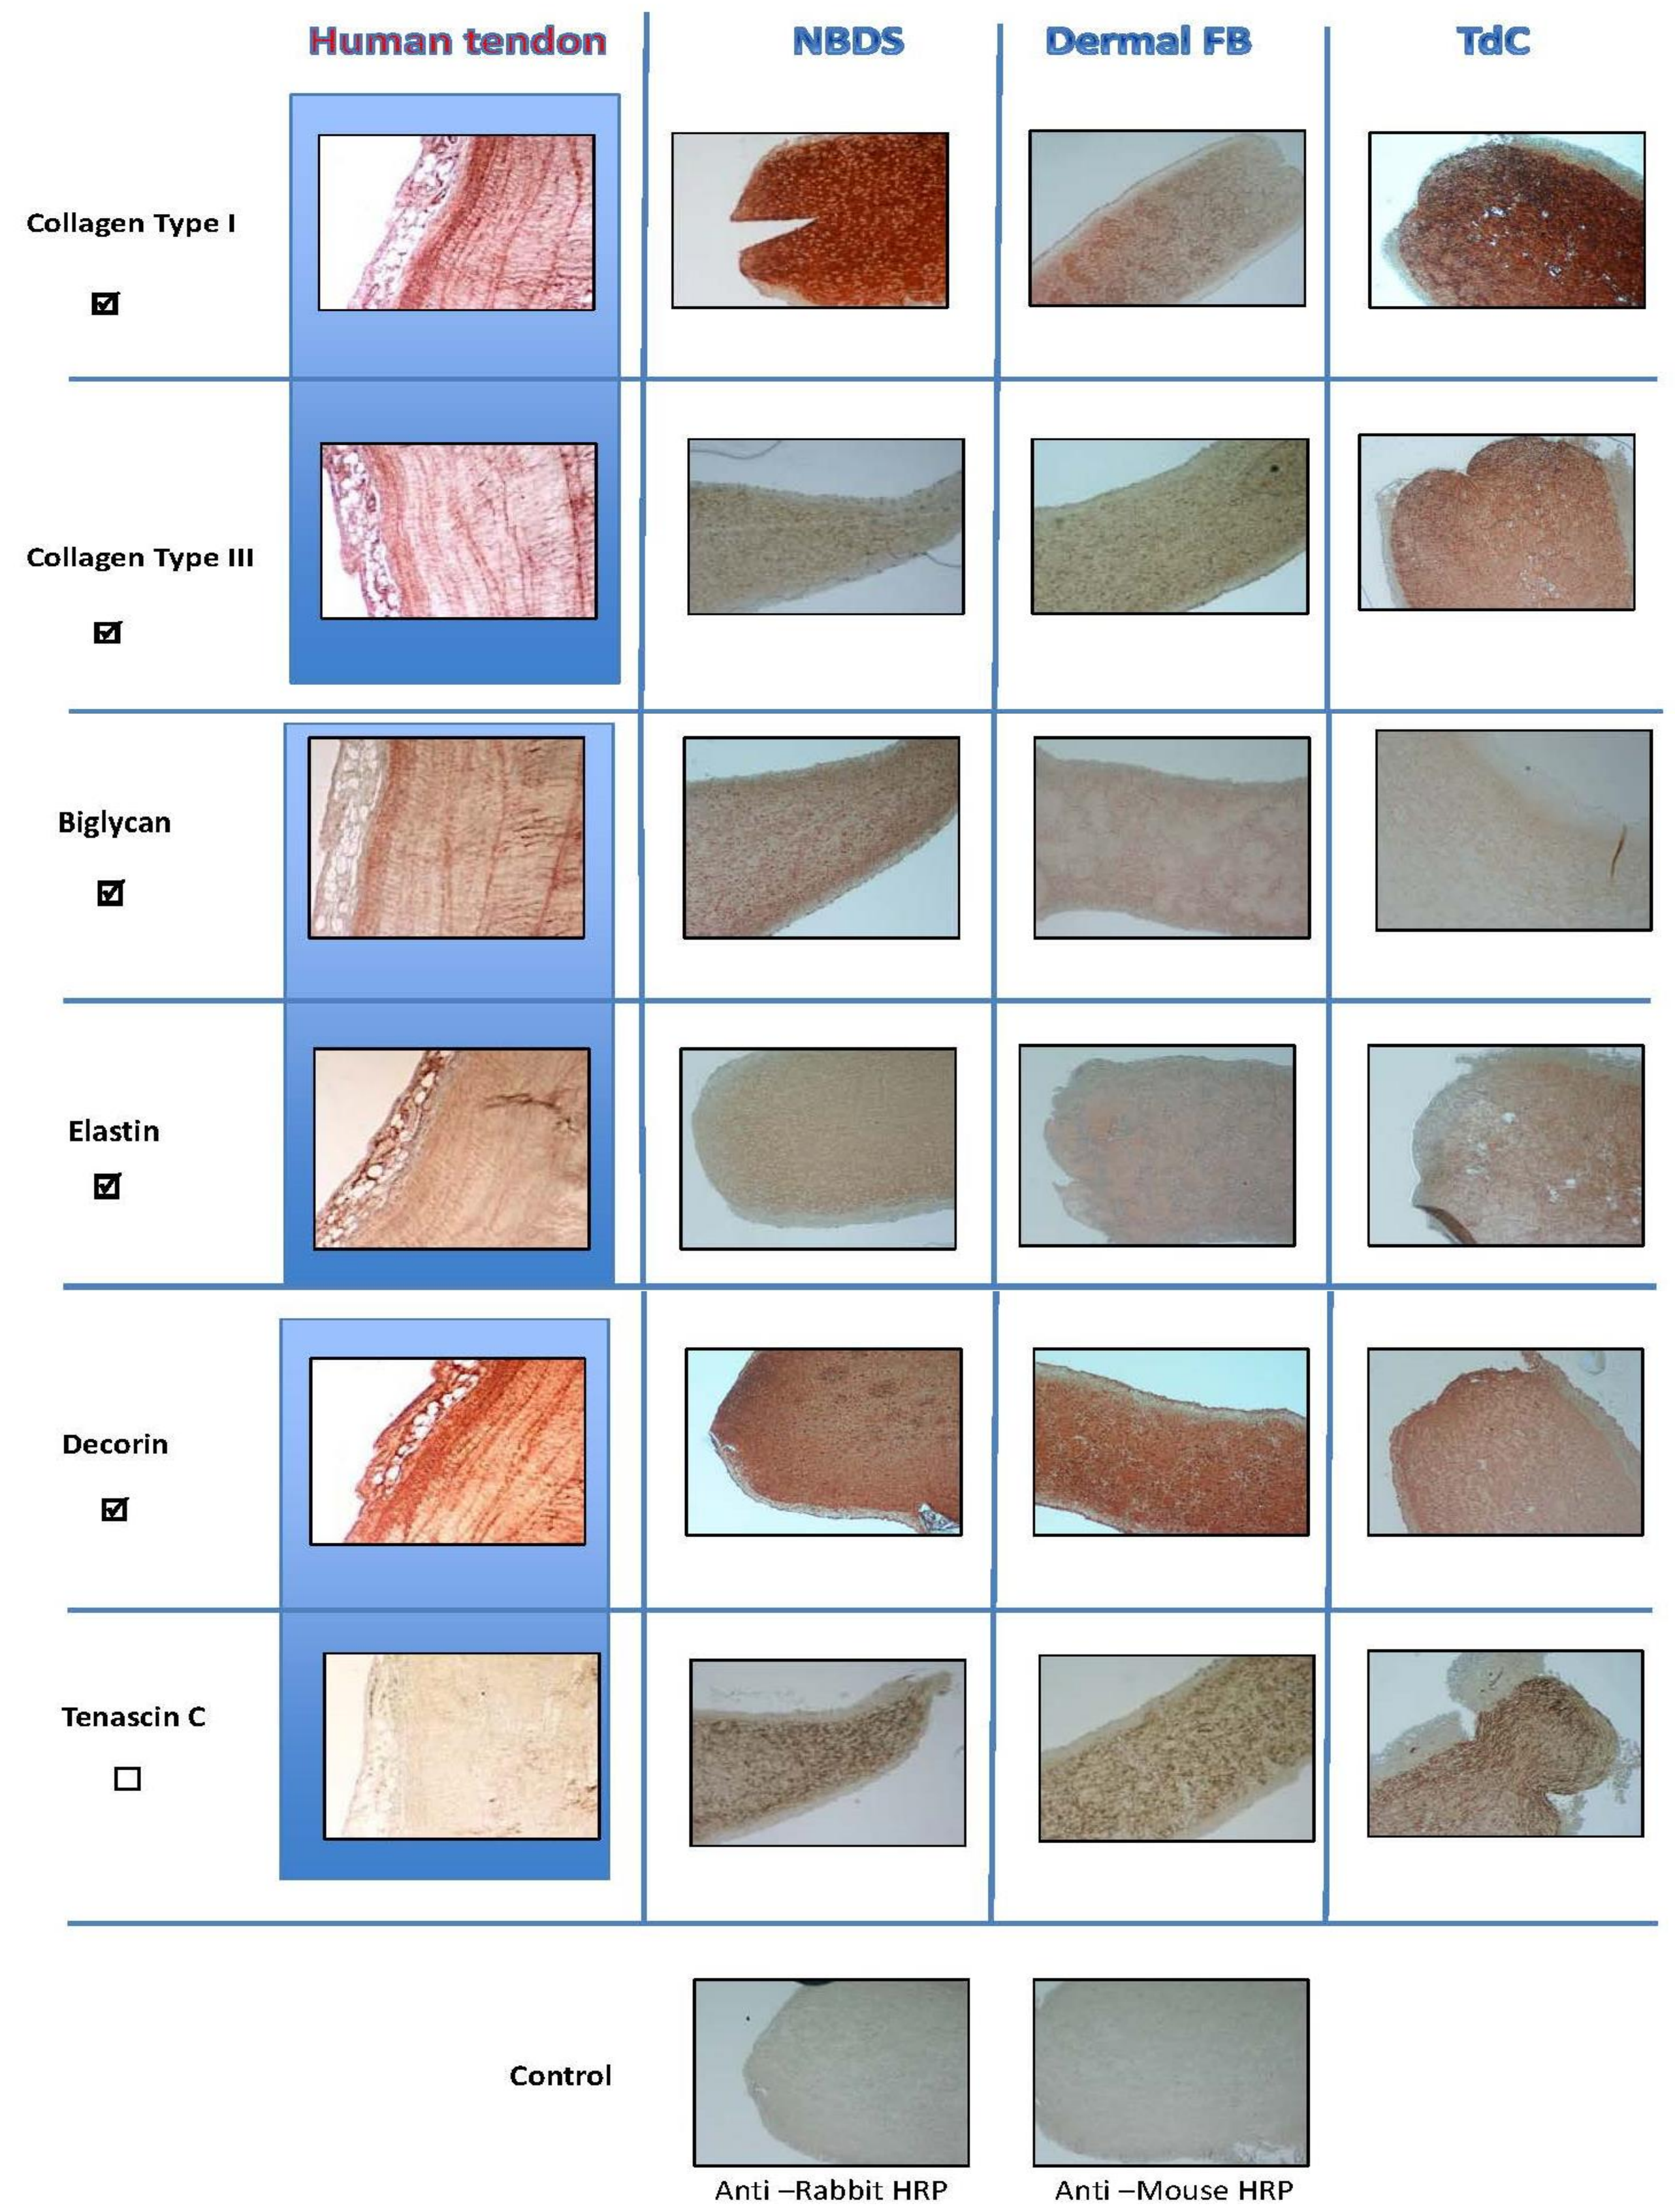

Human tendon
NBDS
Dermal FB
TdC
Collagen Type I
Collagen Type III
Biglycan
Elastin
Decorin
Tenascin C
Control
Anti –Rabbit HRP
Anti –Mouse HRP

Tendon-related protein expression was compared between plasma-embedded NBDS, dermal FB and tendon cells (TdC) after 10-days of linear stretch.

Table 1. Summary of Pre-clinical Safety Studies

In Vivo SAFETY							
Study Purpose	Type	Objectives	Animal Model	Injection Site	Duration	Analysis	Results
Local Tolerance	GLP	To study tolerance of NBDS cells	New Zealand White Rabbits (6)	Intra-tendon: Achilles	5 days	Clinical observation and histopathology of tendon.	No clinical abnormalities or mortality related to treatment: mild reaction at local site in both placebo groups.
Tumorigenicity	GLP	To test potential of NBDS cells to form tumors.	C.B-17 SCID-beige mice (40). Hela adenocarcinoma cells used for positive control.	Sub-cutaneous	3 months	Clinical observation and histopathology.	No abnormal cell growth or tumor formation observed with NBDS cells.
Biodistribution	GLP	To study distribution of NBDS cells post-injection	C.B-17 SCID-beige mice (30).	Intradermal and sub-cutaneous	4 weeks	RT-PCR,	No detection of human cells in all organs and tissues examined.

Conclusions

In summary, our pre-clinical data indicate a safety profile for NBDS cells. NBDS cells will be administered to damaged tendon sites by ultrasound guided injection. Delivering collagen producing cells directly to the site of injury will address the underlying cause of tendinosis and give this therapy unique advantages over currently available treatments that have limited efficacy. Due to an autologous nature of the product, it reduces the likelihood of adverse immune reactions to administration.

Recently, Health Canada did not object for a Phase I/II clinical trial using NBDS cells for the treatment of tendinosis in humans in Canada. The trial is planned to be initiated in 2015.